Poster: A0120

The DAVIO 2 Trial: A Phase 2, Randomized, Double-Masked, Controlled Multicenter Study of EYP-1901 vs Aflibercept in Previously Treated Wet Age-Related Macular Degeneration

Eichenbaum, David A.[1,2]; Hershberger, Vrinda[3]; Patel, Sunil S.[4]; Abbey, Ashkan M.[5]; Bridges, William Z.[6]; Khanani, Arshad M.[7]; Ruggiero, Stephanie[8]; Roy, Monica[8]; Duker, Jay S.[8]; Paggiarino, Dario A.[9]

1. Retina Vitreous Associates of Florida, Saint Petersburg, FL, United States. 2. Morsani College of Medicine, University of South Florida, Tampa, FL, United States. 3. Florida Eye Associates Inc., Melbourne, FL, United States. 4. Ophthalmology Specialists of Texas, PLLC, Abilene, TX, United States. 5. Texas Retina Associates, Dallas, TX, United States. 6. Asheville Eye Associates, Asheville, NC, United States. 7. Sierra Eye Associates and University of Nevada, Reno School of Medicine, Reno, NV, United States. 8. EyePoint Pharmaceuticals, Inc., Watertown, MA, United States. 9. Former employee of EyePoint Pharmaceuticals, Inc., Watertown, MA, United States.

Purpose

- More durable therapies are needed for wet age-related macular degeneration (wAMD).[1-3]
- EYP-1901 is a bioerodible, sustained-delivery, intravitreal (IVT) insert.[1-3]
- Delivers potent pan–vascular endothelial growth factor (VEGF) receptor inhibitor vorolanib.
- DAVIO 2 trial is evaluating the efficacy and safety of EYP-1901 vs aflibercept 2 mg q8W.

Methods

- DAVIO 2 (NCT05381948) is a phase 2, randomized, double-masked, parallel trial of a single EYP-1901 treatment compared to aflibercept 2 mg q8W in previously treated patients with wAMD (**Figure 1**).
- Following the first 3 aflibercept injections, all eyes in all arms were eligible to receive supplemental aflibercept injections if they met certain criteria (**Figure 1**).

Figure 1: Study Design

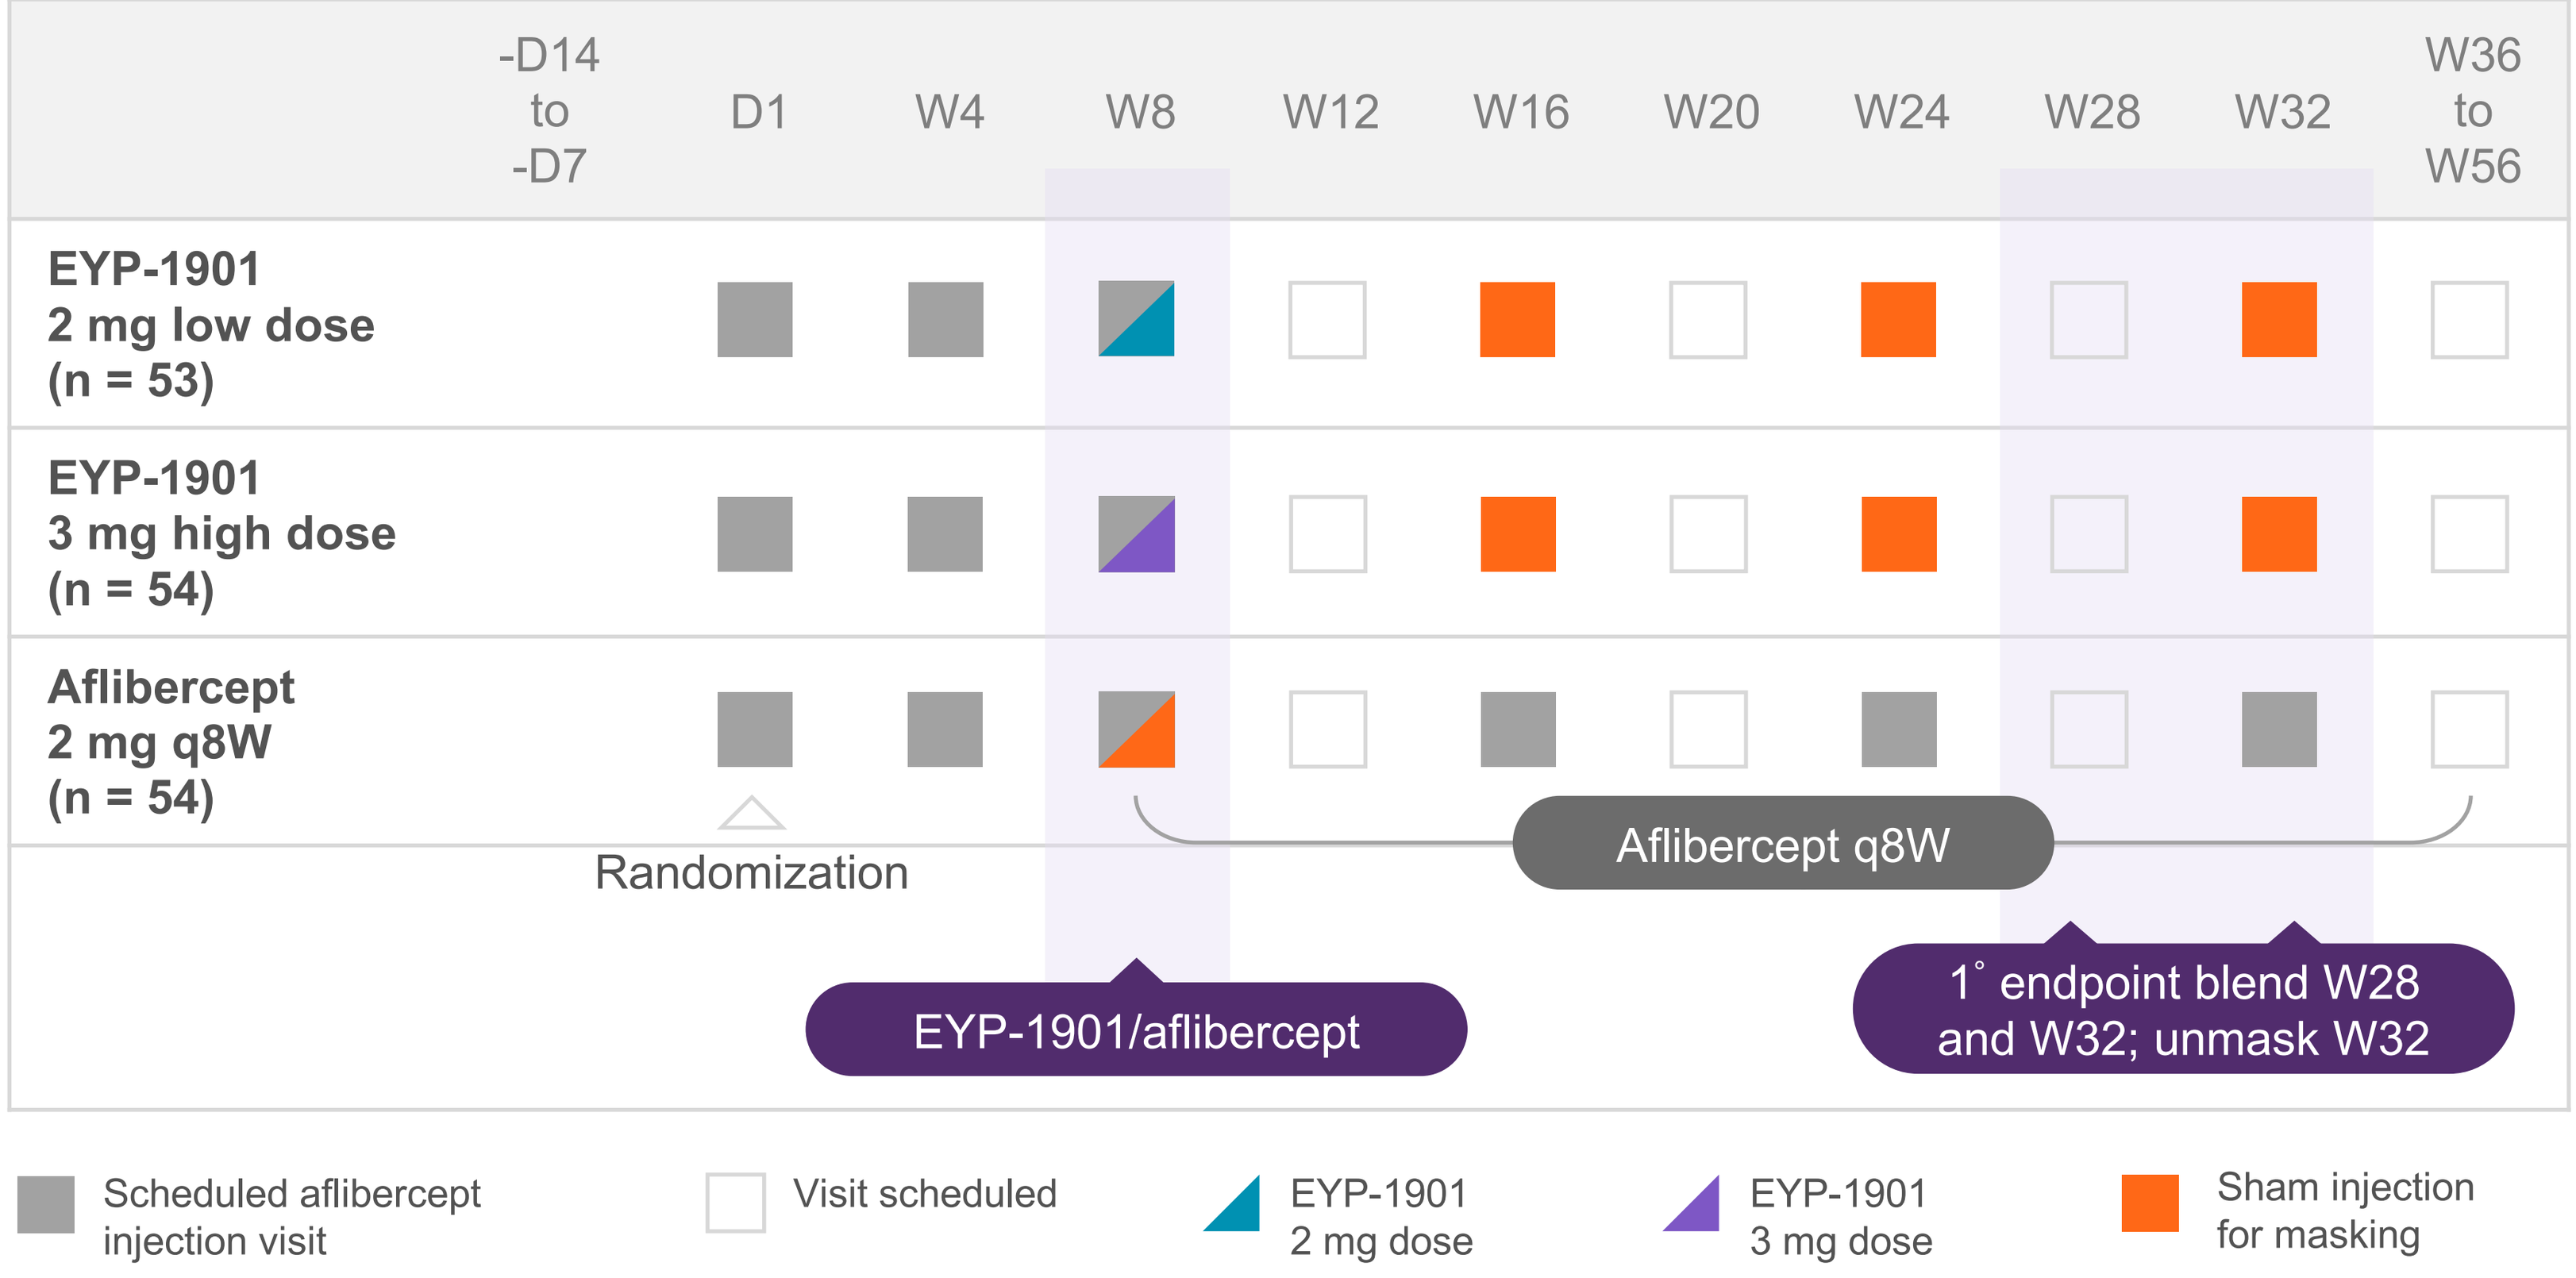

-D14 to -D7
D1
W4
W8
W12
W16
W20
W24
W28
W32
W36 to W56
EYP-1901 2 mg low dose (n = 53)
EYP-1901 3 mg high dose (n = 54)
Aflibercept 2 mg q8W (n = 54)
Randomization
Aflibercept q8W
EYP-1901/aflibercept
1° endpoint blend W28 and W32; unmask W32
Scheduled aflibercept injection visit
Visit scheduled
EYP-1901 2 mg dose
EYP-1901 3 mg dose
Sham injection for masking

Key Inclusion/Exclusion Criteria
- Documented response to anti-VEGF
- ≥2 anti-VEGF injections in previous 6 months
- BCVA range 85 to 35 letters (20/20 to 20/200)
- Excluded CST >350 µm, IRF >25 µm, RPED >400 µm

Criteria for Supplemental Anti-VEGF Therapy

BCVA reduction of ≥5 letters from best on-study measurement due to wAMD AND increase in CST of ≥75 µm from lowest on-study measurement

OR

BCVA reduction of ≥10 letters from best on-study measurement due to wAMD

OR

Increase in CST of ≥100 µm from lowest on-study measurement from 2 consecutive visits

OR

Presence of new or worsening vision-threatening hemorrhage due to wAMD

Primary Endpoint
- Combined mean change in BCVA at Weeks 28 and 32 (6 months after EYP-1901 injection)

Key Secondary Endpoint
- Safety
- Reduction in treatment burden
- Mean change in CST

BCVA, best-corrected visual acuity; CST, central subfield thickness; D, Day; IRF, intraretinal fluid; q8W, every 8 weeks; RPED, retinal pigment epithelium detachment; VEGF, vascular endothelial growth factor; W, Week; wAMD, wet age-related macular degeneration.

Results

Baseline Characteristics

- Baseline characteristics were well balanced across treatment arms.

Primary Endpoint: Mean Change in BCVA

- The primary endpoint was met; both doses of EYP-1901 achieved statistical non-inferiority (**Figure 2A**).
- BCVA in eyes without supplemental injections was similar to the overall population (**Figure 2B**).

Figure 2: Mean Change in BCVA From Baseline Visit

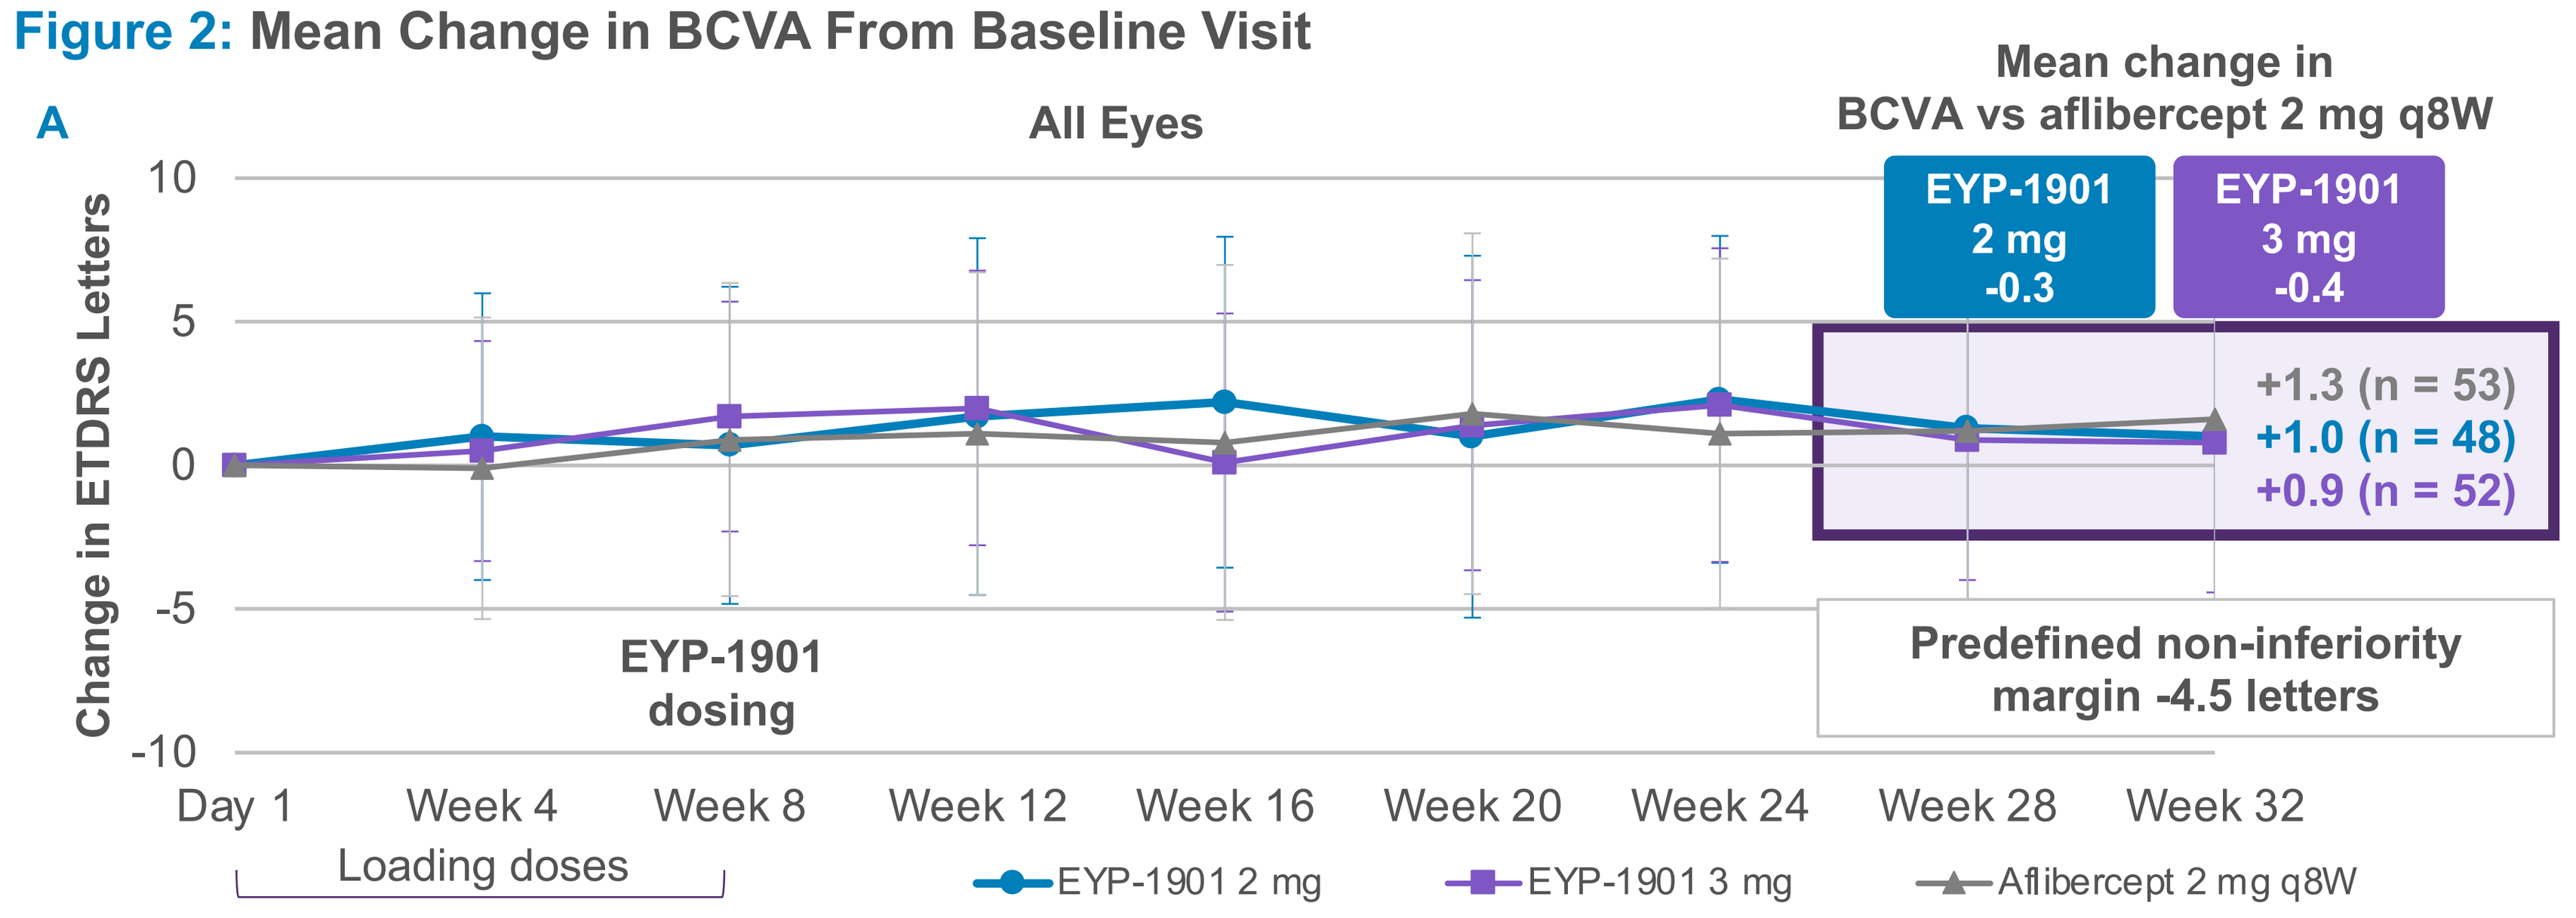

A
All Eyes
Mean change in BCVA vs aflibercept 2 mg q8W
EYP-1901 2 mg -0.3
EYP-1901 3 mg -0.4
+1.3 (n = 53)
+1.0 (n = 48)
+0.9 (n = 52)
Predefined non-inferiority margin -4.5 letters
EYP-1901 dosing
Change in ETDRS Letters
10
5
0
-5
-10
Day 1
Week 4
Week 8
Week 12
Week 16
Week 20
Week 24
Week 28
Week 32
Loading doses
EYP-1901 2 mg
EYP-1901 3 mg
Aflibercept 2 mg q8W

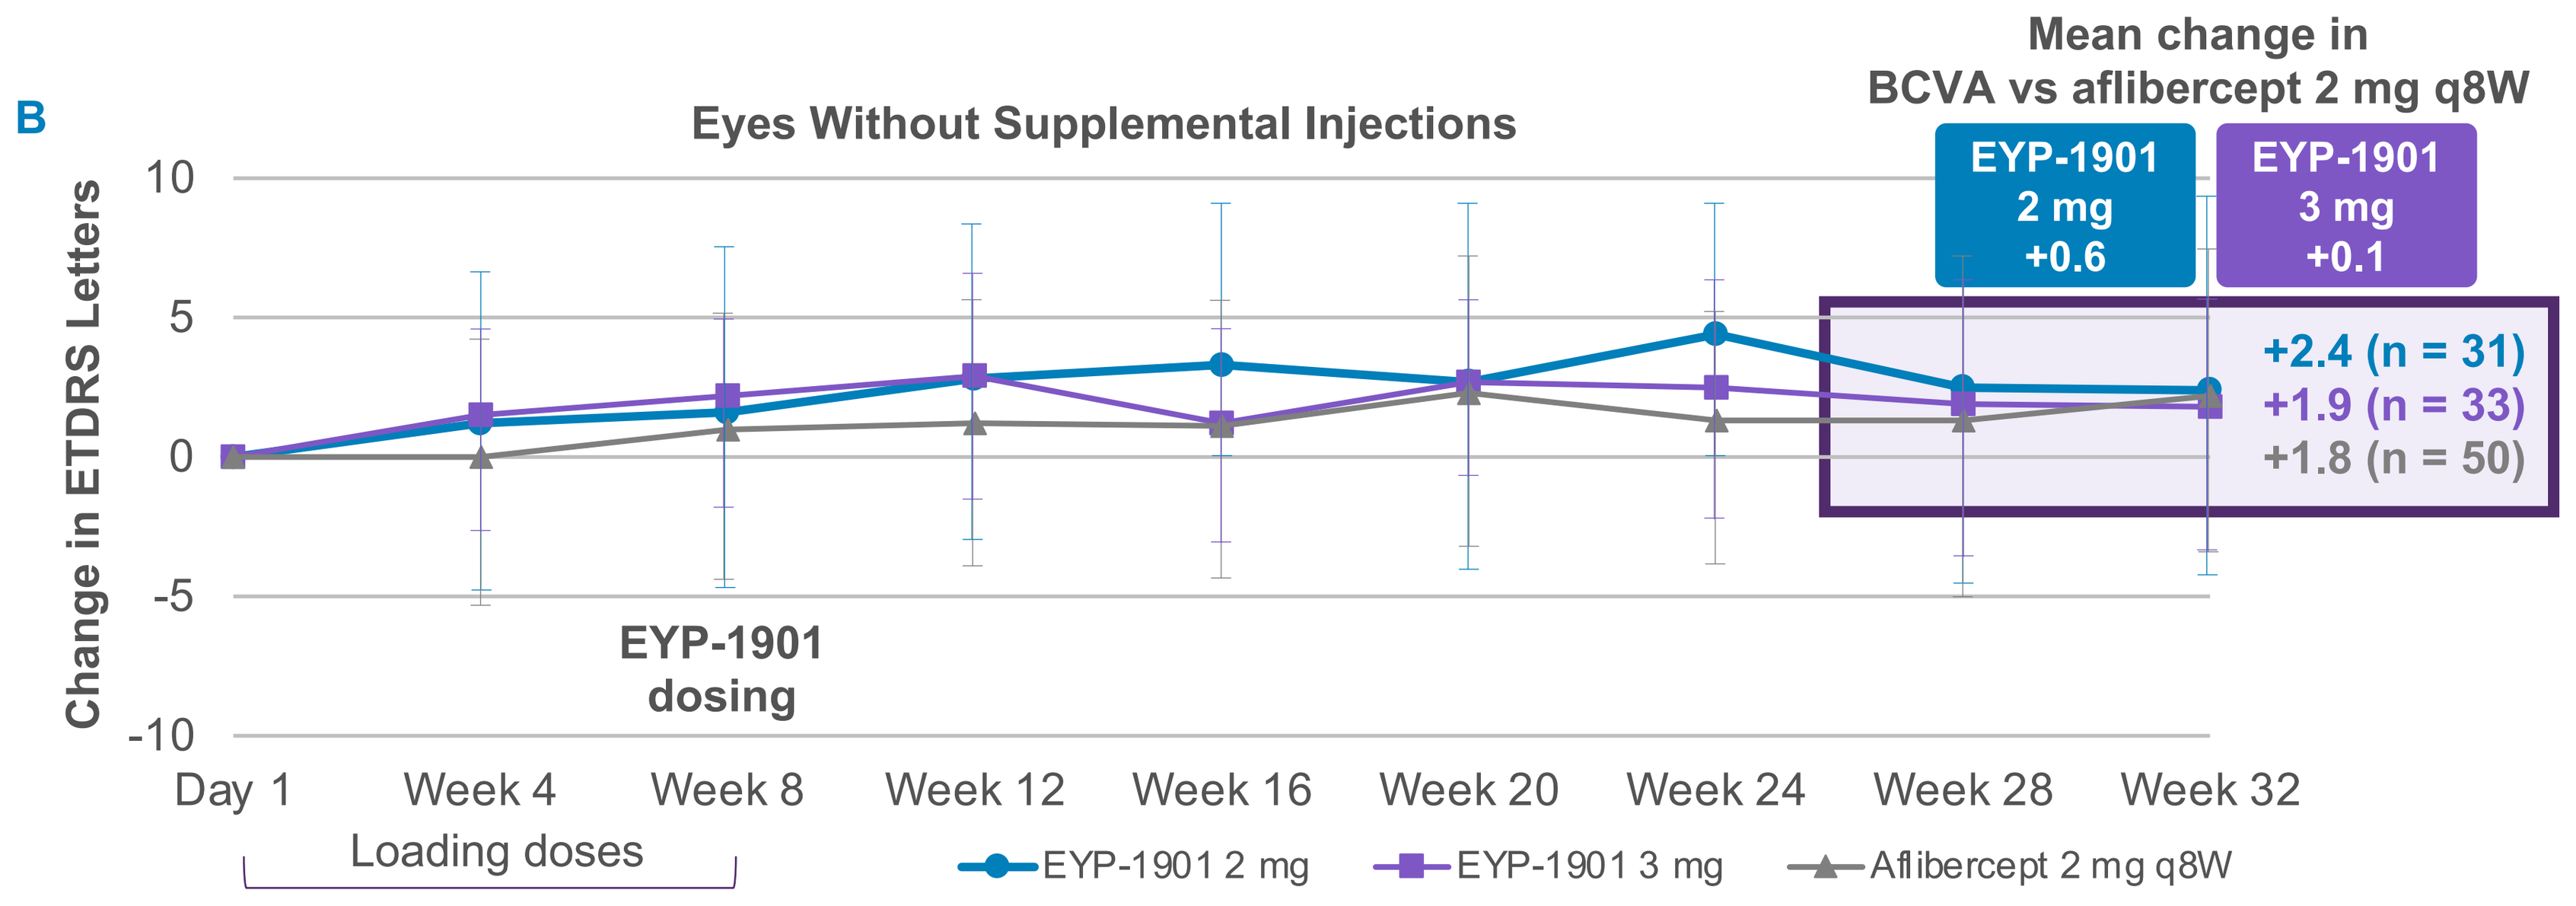

B
Eyes Without Supplemental Injections
Mean change in BCVA vs aflibercept 2 mg q8W
EYP-1901 2 mg +0.6
EYP-1901 3 mg +0.1
+2.4 (n = 31)
+1.9 (n = 33)
+1.8 (n = 50)
EYP-1901 dosing
Change in ETDRS Letters
10
5
0
-5
-10
Day 1
Week 4
Week 8
Week 12
Week 16
Week 20
Week 24
Week 28
Week 32
Loading doses
EYP-1901 2 mg
EYP-1901 3 mg
Aflibercept 2 mg q8W

Error bars represent the standard deviation. BCVA units were ETDRS letters. BCVA, best-corrected visual acuity; ETDRS, Early Treatment Diabetic Retinopathy Study; q8W, every 8 weeks.

Mean Change in CST

- CST with EYP-1901 was similar to aflibercept over 32 weeks, with a mean difference below 10 µm with both doses at Week 32 (**Figure 3**). The sawtooth pattern of CST fluctuation seen with aflibercept was not seen with EYP-1901.

Figure 3: Mean Change in CST From Baseline Visit

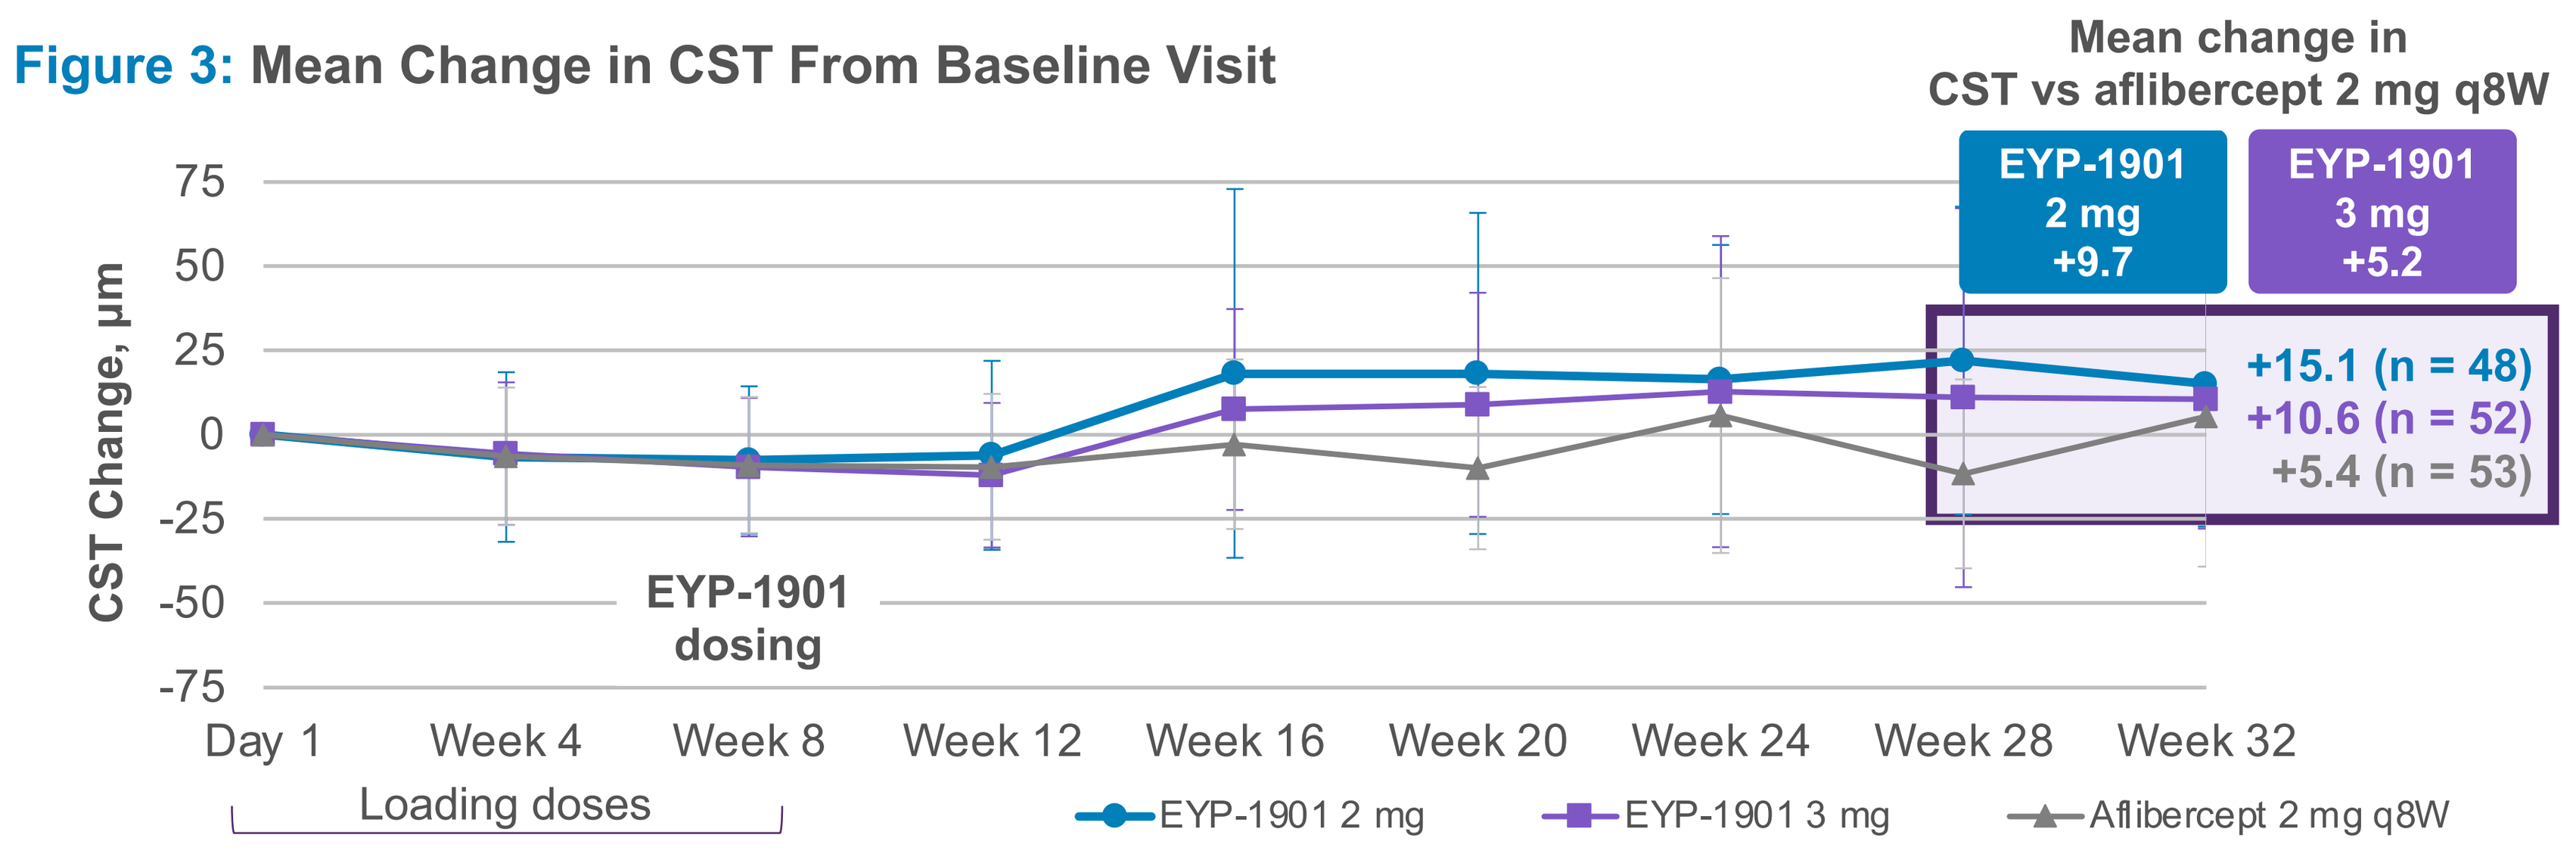

Mean change in CST vs aflibercept 2 mg q8W
EYP-1901 2 mg +9.7
EYP-1901 3 mg +5.2
+15.1 (n = 48)
+10.6 (n = 52)
+5.4 (n = 53)
EYP-1901 dosing
CST Change, µm
75
50
25
0
-25
-50
-75
Day 1
Week 4
Week 8
Week 12
Week 16
Week 20
Week 24
Week 28
Week 32
Loading doses
EYP-1901 2 mg
EYP-1901 3 mg
Aflibercept 2 mg q8W

Error bars represent the standard deviation. CST units were µm. CST, central subfield thickness; q8W, every 8 weeks.

Reduction in Treatment Burden

- 64%–65% of eyes receiving EYP-1901 were supplemental injection–free up to Week 32 (**Figure 4**).

Figure 4: Eyes Supplemental Injection–Free up to Week 32

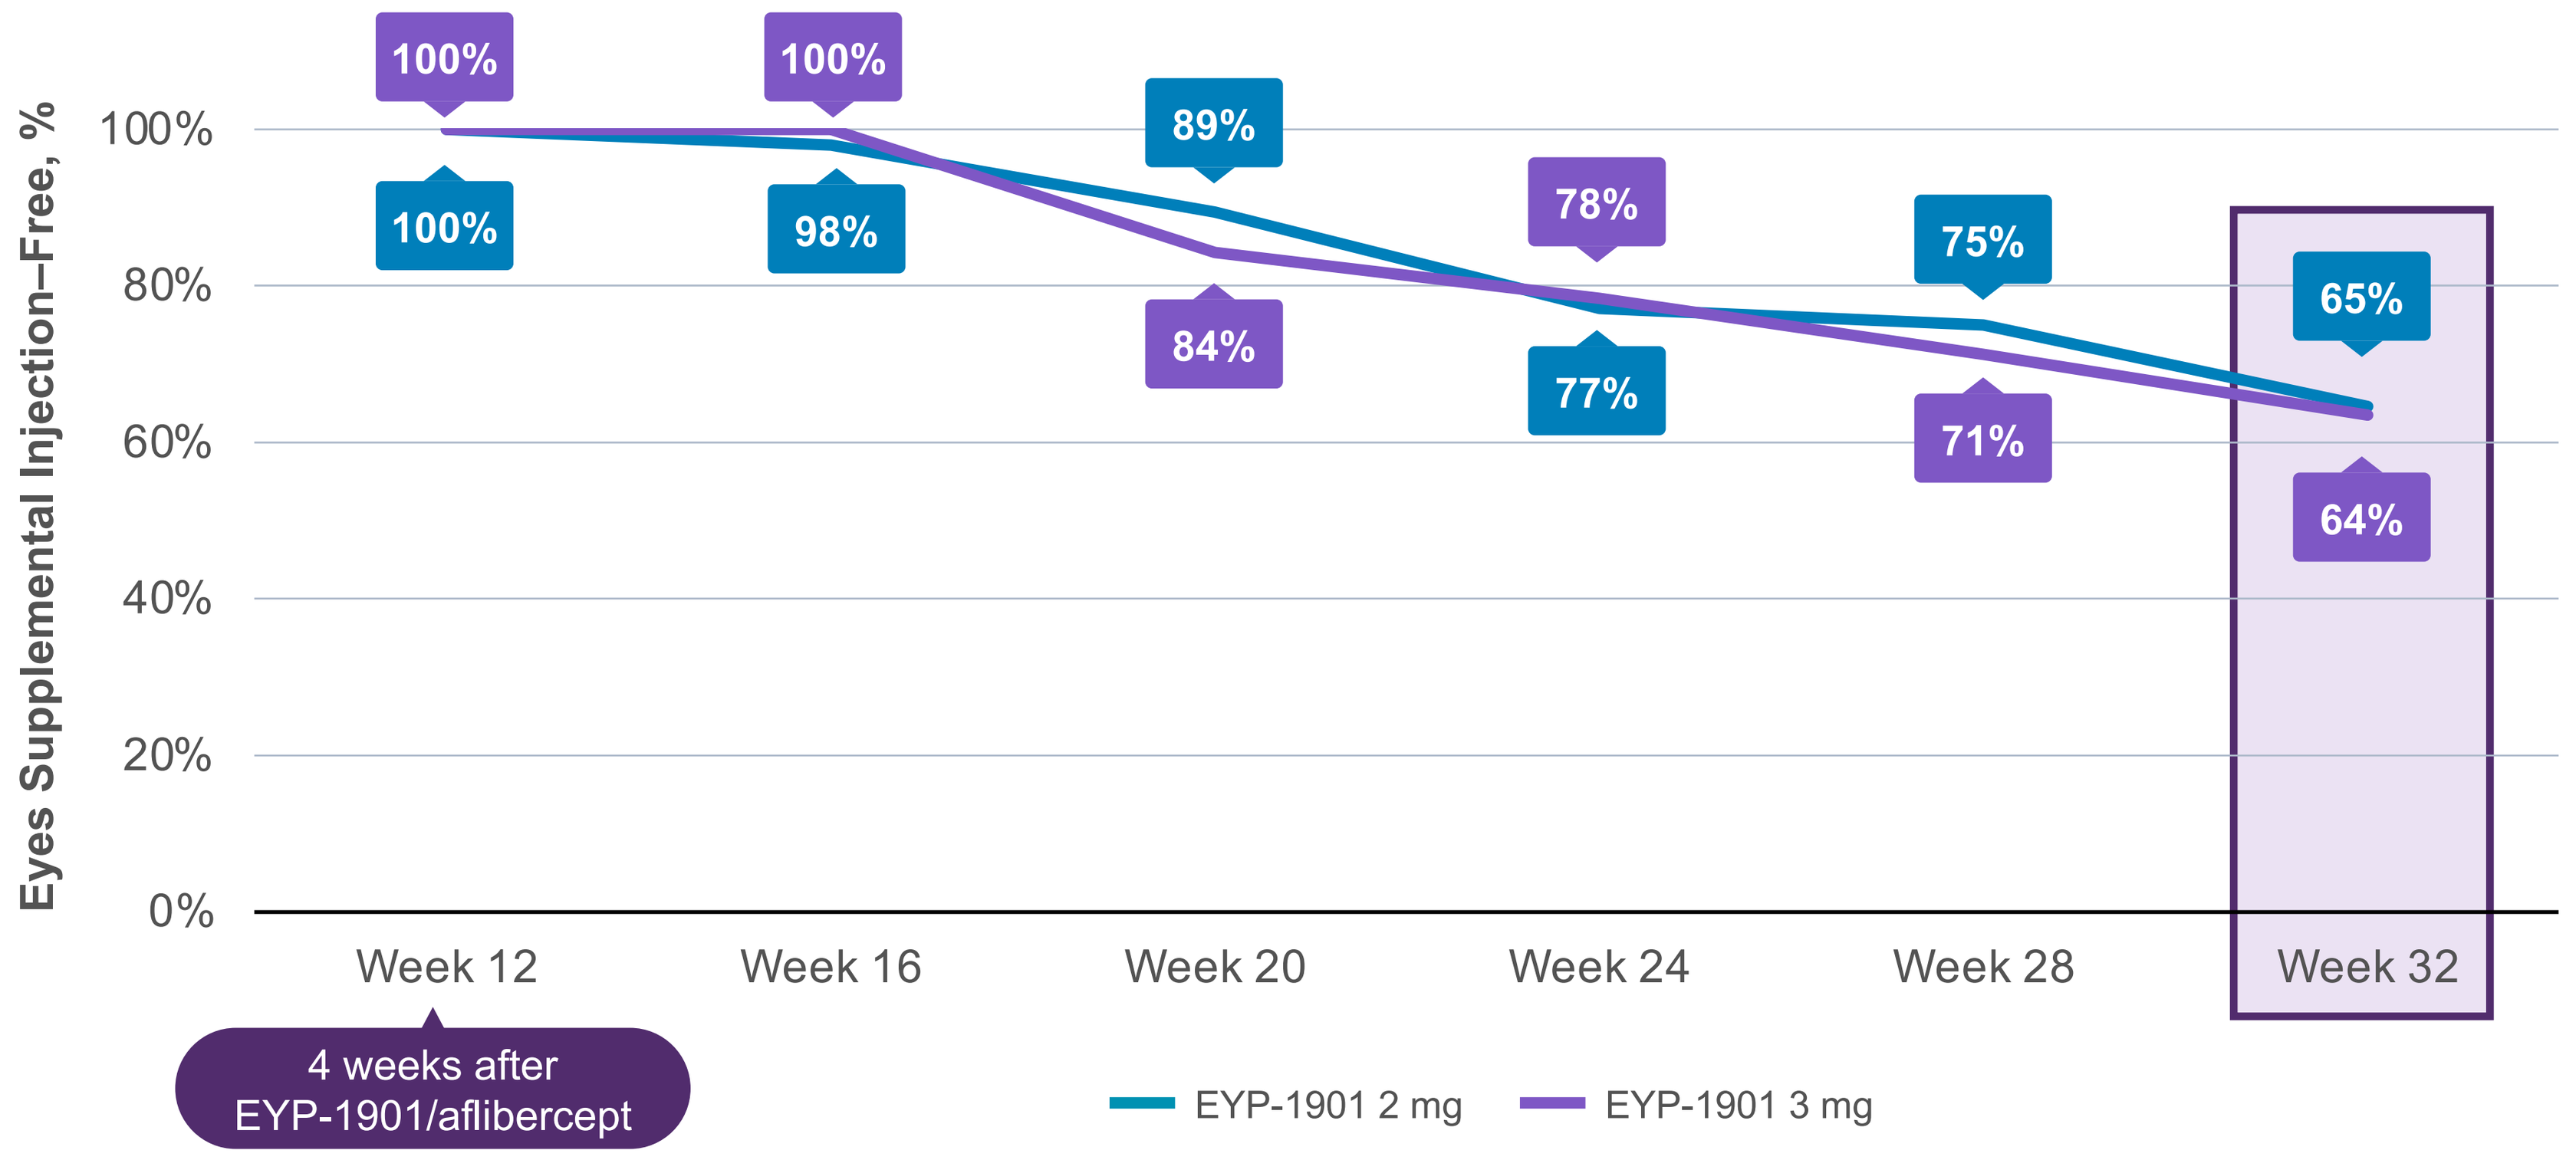

Eyes Supplemental Injection–Free, %
100%
80%
60%
40%
20%
0%
Week 12
Week 16
Week 20
Week 24
Week 28
Week 32
100%
100%
100%
98%
89%
84%
78%
77%
75%
71%
65%
64%
4 weeks after EYP-1901/aflibercept
EYP-1901 2 mg
EYP-1901 3 mg

- There was a reduction in treatment burden of 85%–89% following EYP-1901 treatment, up to Week 32 (**Figure 5**).

Figure 5: Treatment Burden on Study (up to Week 32) vs Prior 6 Months

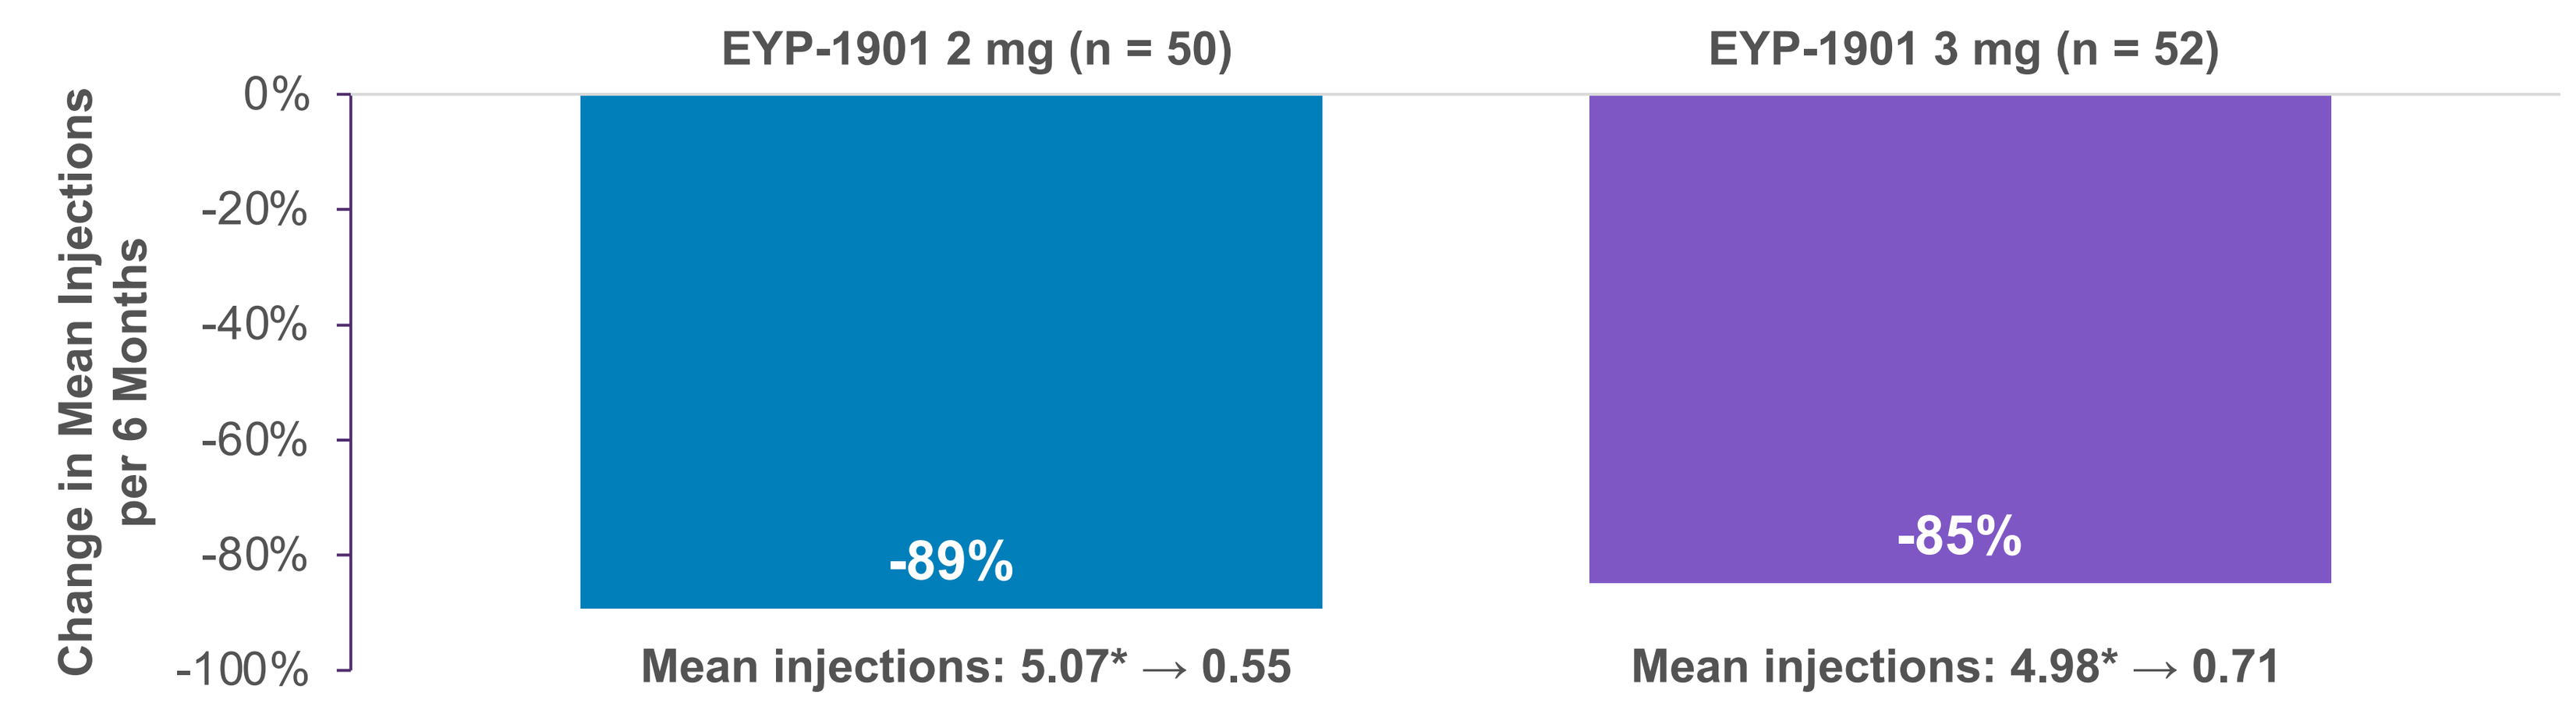

EYP-1901 2 mg (n = 50)
EYP-1901 3 mg (n = 52)
Change in Mean Injections per 6 Months
0%
-20%
-40%
-60%
-80%
-100%
-89%
-85%
Mean injections: 5.07* → 0.55
Mean injections: 4.98* → 0.71

*Normalized.

Safety

- EYP-1901 was well tolerated through 32 weeks with no ocular serious adverse events (AEs) related to EYP-1901. Four serious ocular AEs were reported in study eyes and deemed unrelated to EYP-1901.
- More than 97% of AEs were mild and generally expected with IVT injections. There were no cases of insert migration into the anterior chamber or retinal occlusive vasculitis and no discontinuations related to treatment with EYP-1901.

Conclusions

- EYP-1901 demonstrated statistical non-inferiority to aflibercept with stable BCVA, strong anatomical control, a favorable safety profile, and durability up to 6 months in eyes with wAMD.
- ≥64% of eyes receiving EYP-1901 remained supplemental injection–free up to 6 months, and there was an ≥85% mean reduction in treatment burden vs the prior 6 months.
- Most ocular AEs reported were expected with IVT injection, and no ocular serious AEs were related to EYP-1901.
- 12-month data for DAVIO 2 will be available in the second half of 2024. EYP-1901 is also being studied in phase 2 trials in non-proliferative diabetic retinopathy and diabetic macular edema. A phase 3 pivotal trial in wAMD will begin in the second half of 2024.

References: 1. Thomas CN, et al. *Br J Pharmacol.* 2022;179:1908-1937. 2. Sobolewska B, et al. *Clin Ophthalmol.* 2021;15:4317-4326. 3. Patel P, et al. *J Clin Med.* 2021;10:2436.

Commercial Relationships Disclosure: D.A.E: Financial Support – 4DMT, Alexion, Alimera, Alkahest, Allegenesis, Allergan, Annexon, Apellis, AsclepiX, Bausch & Lomb, Bayer, Chengdu, Coherus, Crinetics, DORC, EyePoint, Gemini, Genentech, Gyroscope, Ionis, Iveric Bio, KKR, Kodiak Sciences, Mylan, NGM, Novartis, Ocular Therapeutix, Opthea, Outlook, Recens Medical, Regeneron, RegenXBio, ReVive, Unity Biotechnology, US Retina; Non-remunerative – Boston Image Reading Center, Hemera Biopharmaceuticals, Network Eye, ReVive, US Retina. **V.H.:** Consultant/Contractor – Genentech. **S.S.P.:** Consultant/Contractor – AiViva, Allergan, Allgenesis, Genentech-Roche, Kala, Kodiak Sciences, Ocugenix, RegenXBio; Financial Support – Aerie, Aerpio, Allergan, Allgenesis, Apellis, Boehringer Ingelheim, Chengdu Kanghong, Clearside, EyePoint, Genentech-Roche, Ionis Pharmaceuticals, Iveric Bio, KalVista, Kodiak Sciences, Mylan, Novartis, Oculis, Opthea, Ora, Oxurion, Regeneron, Samsung, SmileBiotek, Stealth Biotherapeutics, ThromboGenics, Xbrane Biopharma; Employment – Allgenesis Biotherapeutics, Inc. **A.M.A.:** Consultant/Contractor – EyePoint Pharmaceuticals. **W.Z.B.:** No commercial relationships. **A.M.K.:** Consultant/Contractor – Alcon Laboratories, Alimera Sciences, Allergan, EyePoint, Genentech, Outlook Therapeutics, RecensMedical, Regeneron Pharmaceuticals; Recipient – Alimera, Allergan, Biogen MA, Genentech, Regeneron Pharmaceuticals; Personal Financial Interest – Outlook Therapeutics; Financial Support – Allergan, EyePoint. **S.R., M.R., J.S.D.:** Employment – EyePoint Pharmaceuticals. **D.A.P.:** Former employee of EyePoint Pharmaceuticals. **Acknowledgments:** Medical writing services were provided by Scott Salsman, PhD, of Two Labs Pharma Services.

Presented at ARVO 2024 Annual Meeting, May 5–9, 2024; Seattle, WA.

Please visit eyepointpharma.com for more information.